New Use of Niacin for the Treatment of NASH/Non-Alcoholic Fatty Liver Disease (NAFLD): combination therapy with drugs in development

Moti L. Kashyap, Vaijinath (Vijay) S. Kamanna

Aasta Pharmaceuticals, LLC, Irvine, California

ABSTRACT

In pharmacologic doses, niacin has beneficial effects on dyslipidemia. As monotherapy, it prevents heart attacks and stroke.

In preclinical research, Niacin inhibits and reverses hepatic steatosis, inflammation and fibrosis by unique mechanisms including reduction of oxidative stress, and inhibition of DGAT2.

This data indicates a new use of an old drug used clinically for dyslipidemia.

A clinical trial in 39 dyslipidemic patients with steatosis showed a statistically significant reduction of liver fat by 47% and significant reductions in liver enzymes and C-Reactive Protein (CRP) when treated with Extended-Release Niacin (generic for FDA approved Niaspan) for 6 months.

Because niacin acts on all 3 major stages of NASH-NAFLD, combination with a drug in development could result in a broader and more intense efficacy than either drug alone.

The novel repurposed patented use of Niacin for Fatty Liver Disease (1) gives a tremendous time lead in drug development for NASH. It will also treat dyslipidemia which is often found in the same NASH patient.

Niacin-based combination therapy research may produce one of the most effective broad-spectrum products for NASH.

BACKGROUND

- **Nonalcoholic fatty liver disease (NAFLD) is the leading cause of chronic liver disease and significantly contributes to liver-related morbidity and mortality, and affects about 30% of US population (2, 3).**
- **Non-alcoholic steatohepatitis (NASH) is an aggressive subtype of NAFLD defined by the presence of steatosis (fatty liver), inflammation and hepatocellular ballooning with or without fibrosis (4-6).**
- **In USA, 20-25 million people are affected by NASH. NASH with fibrosis is a major complication in about 20% of these patients and leads to the progression to cirrhosis, hepatocellular cancer and liver failure resulting in death (4-6).**
- **It is well known that NASH-NAFLD, Dyslipidemia and Atherosclerotic Cardiovascular Disease often occur in the same patient but this is not fully recognized in clinical practice.**
- **In pharmacologic doses, niacin is used for dyslipidemia therapy. It prevents heart attacks and stroke. In combination with statins, it effects reversal of atherosclerosis in coronary, carotid and femoral arteries in patients with dyslipidemia.**
- **In recent evidence described in this presentation, we show that niacin has powerful effects on all major stages of NAFLD-NASH.**
- **Thus, niacin uniquely offers the potential of not only treating NASH but also dyslipidemia which are often found in the same NASH patient.**

METHODS – Pre-Clinical Studies

- In-Vitro Studies: Human primary hepatocytes (Lonza) or human hepatoblastoma cell line HepG2 cells (ATCC) were used to examine the effect of niacin on fat accumulation, DGAT2 mRNA expression, and ROS production.
- In-Vitro Studies: Human primary hepatic Stellate cells from normal subjects and from NASH subjects (Samsara Sciences, San Diego, CA) were used to assess collagen type 1 deposition as a measure of fibrosis.
- In-Vivo Studies:
 a) Prevention: Rats were fed either a rodent normal chow, chow containing high-fat (HF), or HF containing 0.5% or 1% niacin in the diet for 4 weeks.
 b) Regression: Rats were first fed HF diet for 6 weeks to induce hepatic steatosis, and then treated with niacin (0.5% in the diet) while continuing on HF diet for 6 weeks.
- Hepatocyte fat accumulation: Staining with Nile Red O and measurement of fluorescence. Triglyceride content was measured using an assay kit.
- Hepatocyte and Stellate Cell Reactive Oxygen Species (ROS): Staining with DCFDA and measurement of fluorescence. Fluorescence intensity units were used as an index of ROS production. Interleukin -8 (IL-8) was measured by ELISA.
- Stellate cell fibrosis: Collagen type1 measured by ELISA and by staining total collagen with Sirus Red.
- Liver histology: Hepatic steatosis assessed in H&E stained liver sections.

Niacin Prevents and Regresses Hepatocyte Fat Content and Steatosis in Rat Model via DGAT2 Inhibition

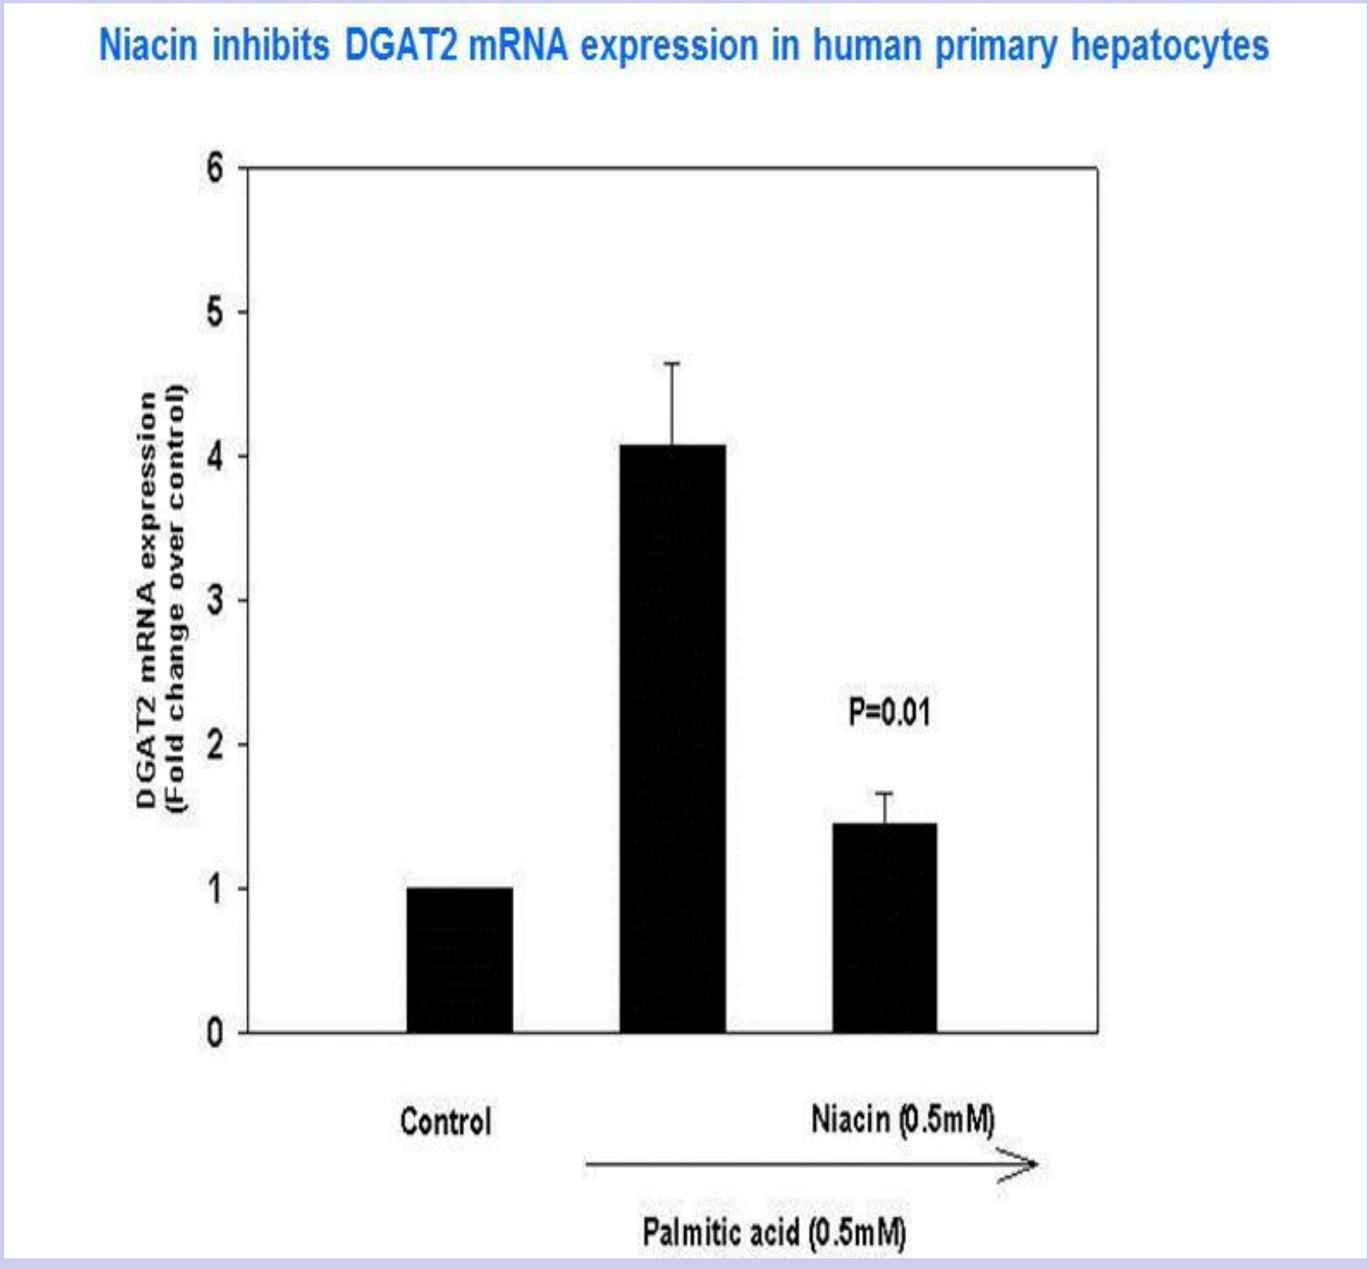
Niacin inhibits DGAT2 mRNA expression in human primary hepatocytes
DGAT2 mRNA expression (Fold change over control)
P=0.01
Control
Niacin (0.5mM)
Palmitic acid (0.5mM)

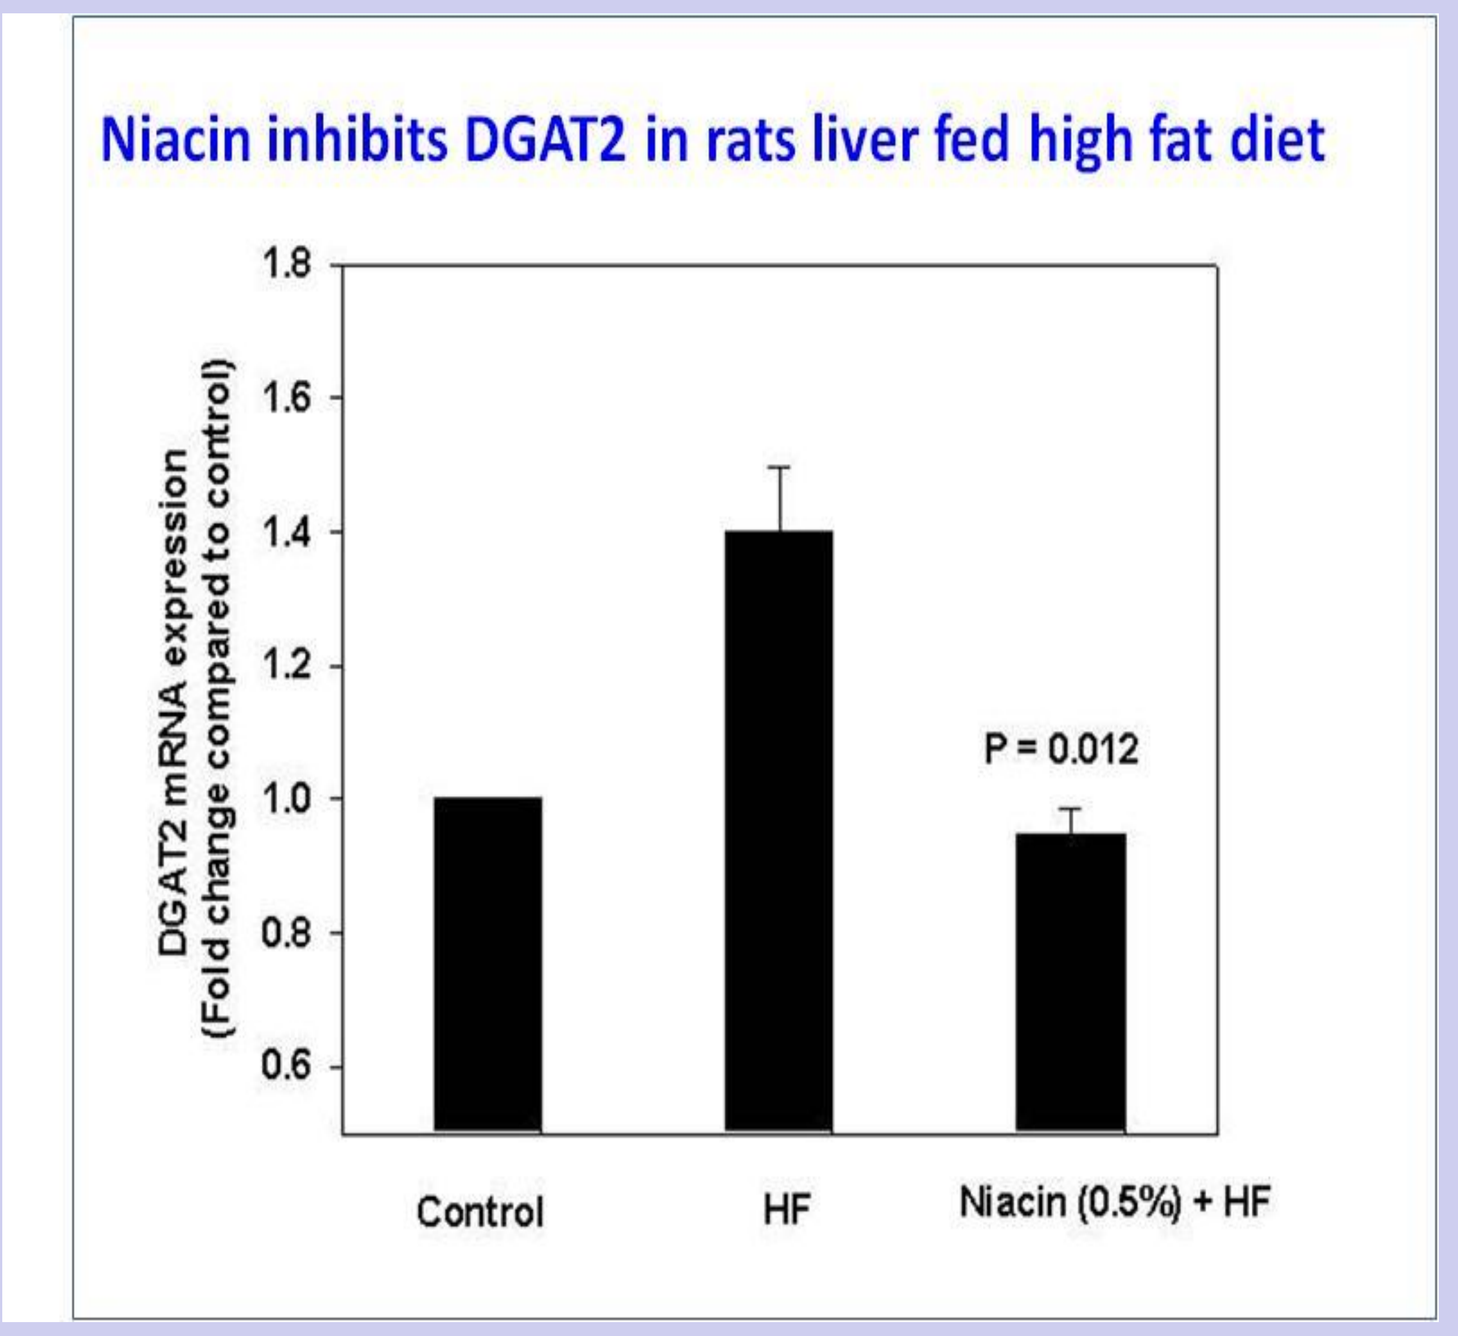
Niacin inhibits DGAT2 in rats liver fed high fat diet
DGAT2 mRNA expression (Fold change compared to control)
P = 0.012
Control
HF
Niacin (0.5%) + HF

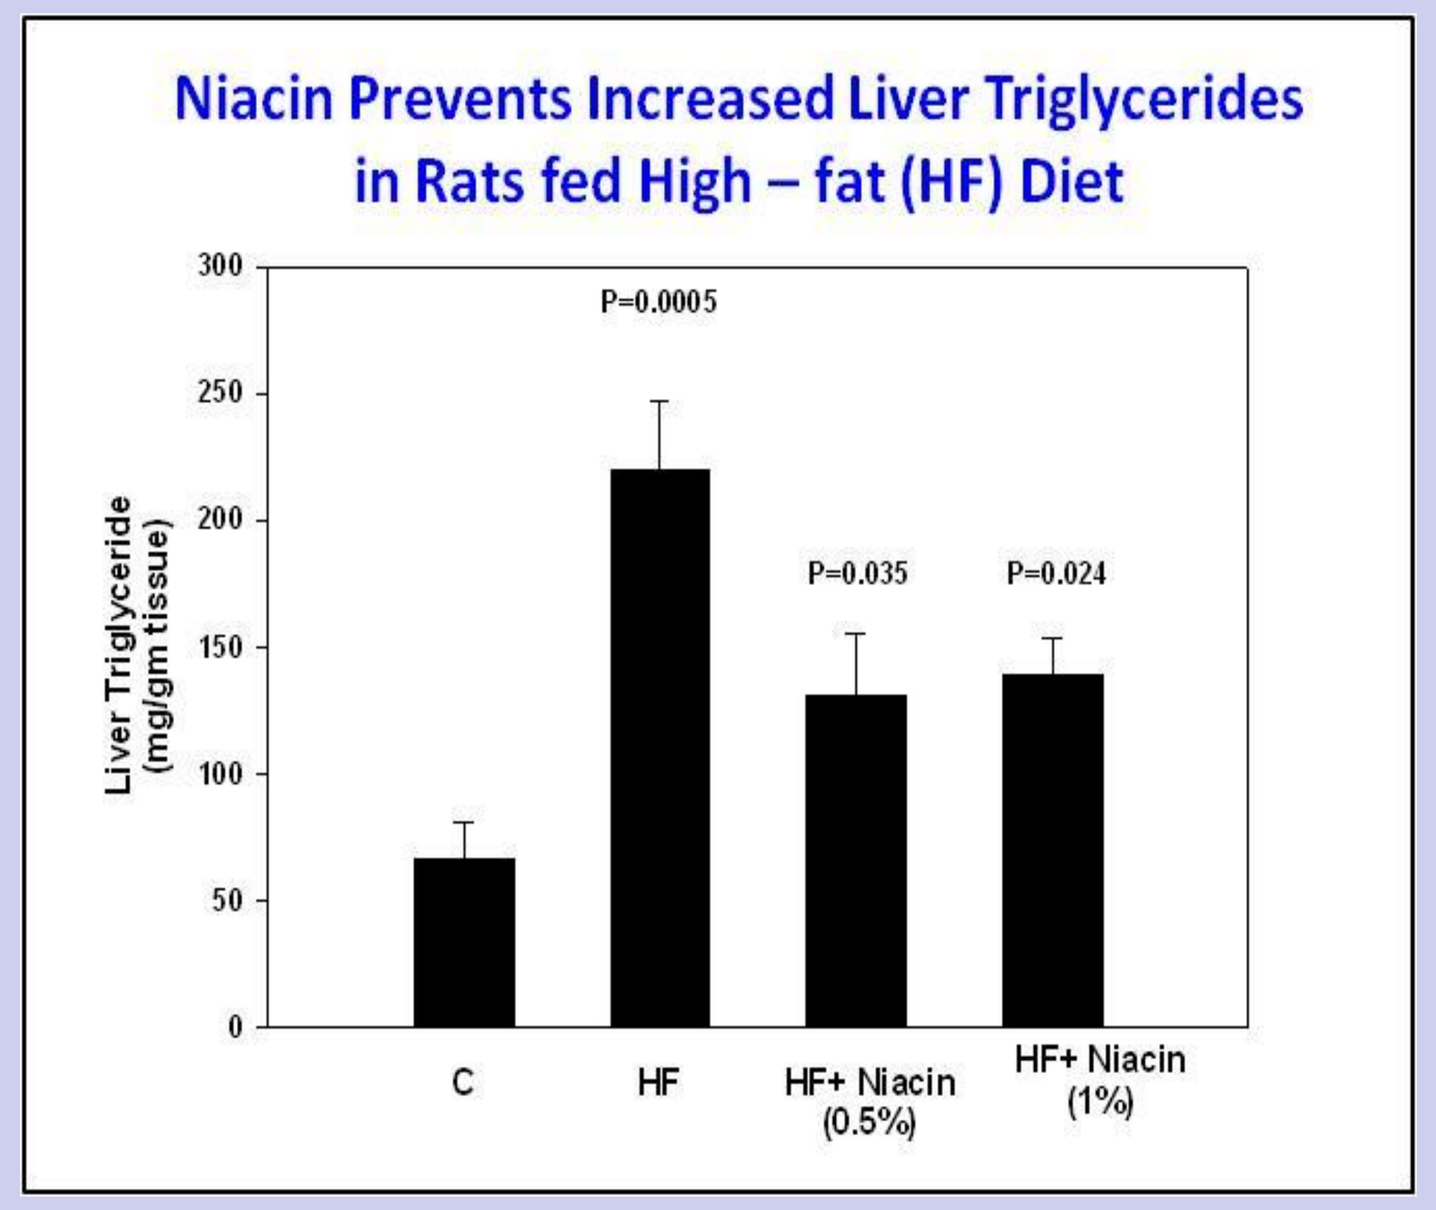
Niacin Prevents Increased Liver Triglycerides in Rats fed High – fat (HF) Diet
Liver Triglyceride (mg/gm tissue)
P=0.0005
P=0.035
P=0.024
C
HF
HF+ Niacin (0.5%)
HF+ Niacin (1%)

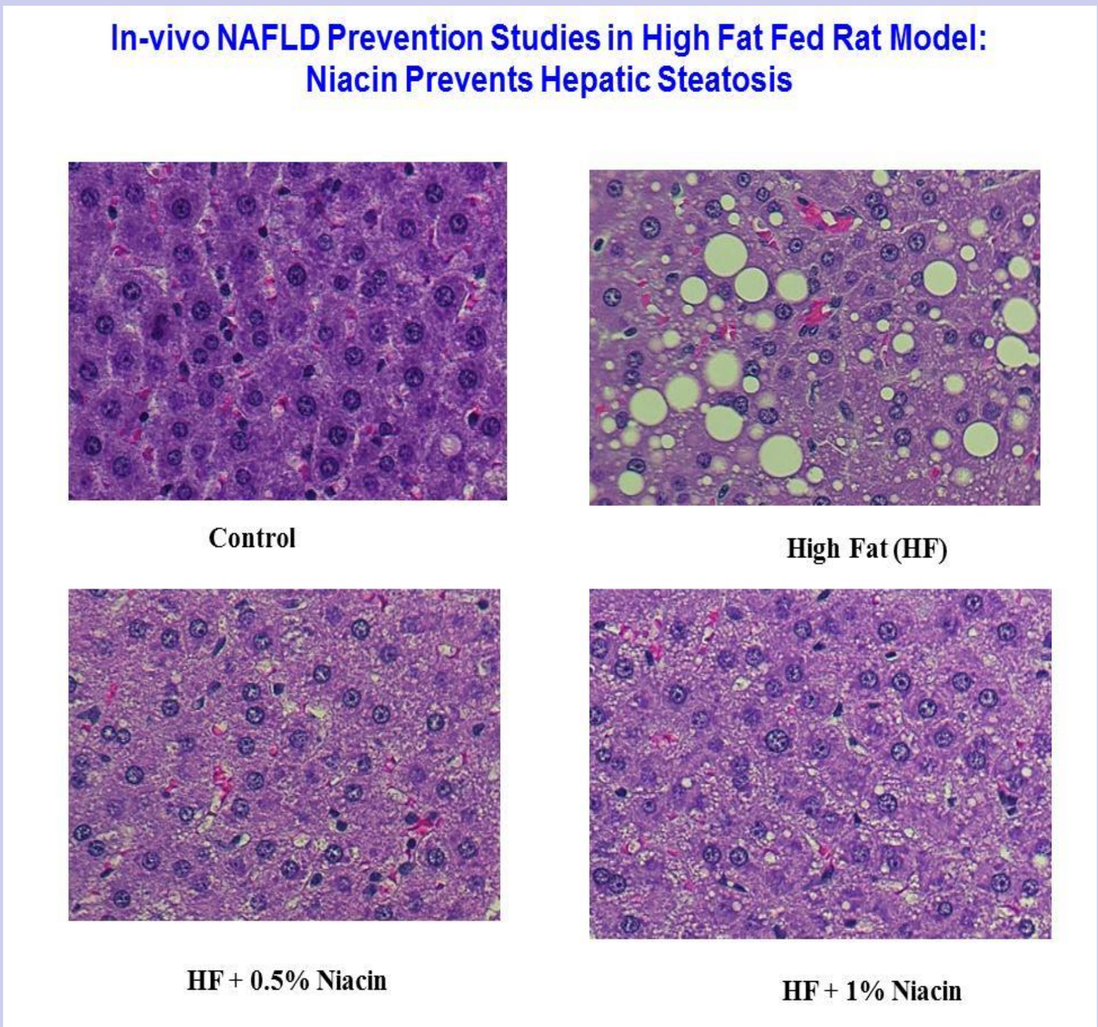
In-vivo NAFLD Prevention Studies in High Fat Fed Rat Model: Niacin Prevents Hepatic Steatosis
Control
High Fat (HF)
HF + 0.5% Niacin
HF + 1% Niacin

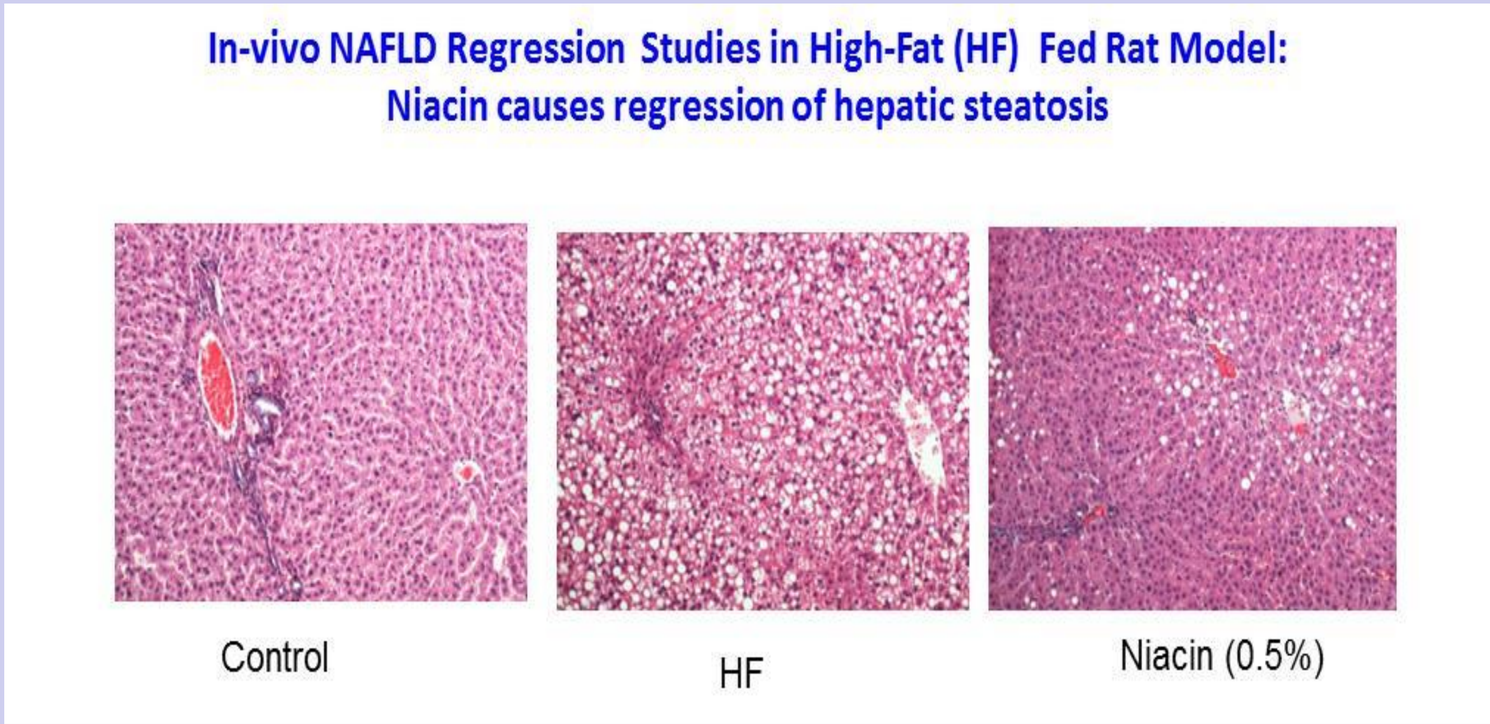
In-vivo NAFLD Regression Studies in High-Fat (HF) Fed Rat Model: Niacin causes regression of hepatic steatosis
Control
HF
Niacin (0.5%)

1. Ganji SH, et.al. Am J Physiol Gastrointest Liver Physiol 306: G320-G327, 2014.
2. Ganji SH,et.al. Metabolism 64:980-990, 2015

Clinical Trial Evidence:
Niacin decreases fatty liver and improves liver enzymes
Hu M., et al. J Lipid Res. 53:802-809, 2012

Study Design/Methods:
- Hypertriglyceridemic patients (n=39, baseline liver fat content 12.8±7.6% were treated with extended-release niacin (2g/day) for 23 weeks
- Liver fat content before and after niacin treatment was measured by Proton MR spectroscopy. Uncontrolled trial.

Results:

	Before	After Treatment
Liver fat content (%)	12.8 ± 7.6	6.7 ± 6.1 (-47.2%, p<0.001)
Fatty liver (liver fat >5%)	31 pts.(79.5%)	19 pts.(48.7%), p<0.01
ALT (IU/l)	67.2 ± 15.7	63.0 ± 16.0 (p<0.026)
Alkaline phosphatase (U/l)	33.0 ± 12.5	28.5 ± 11.6 (p<0.003)
Gamma-glutamyltransferase (U/l)	41.6 ± 18.2	37.1 ± 26.6 (p<0.019)
hsCRP (mg/l)	0.32 ± 0.32	0.25 ± 0.27 (p<0.043)

Niacin Reduces Oxidative Stress/Inflammation in Human Hepatocytes and Stellate cells

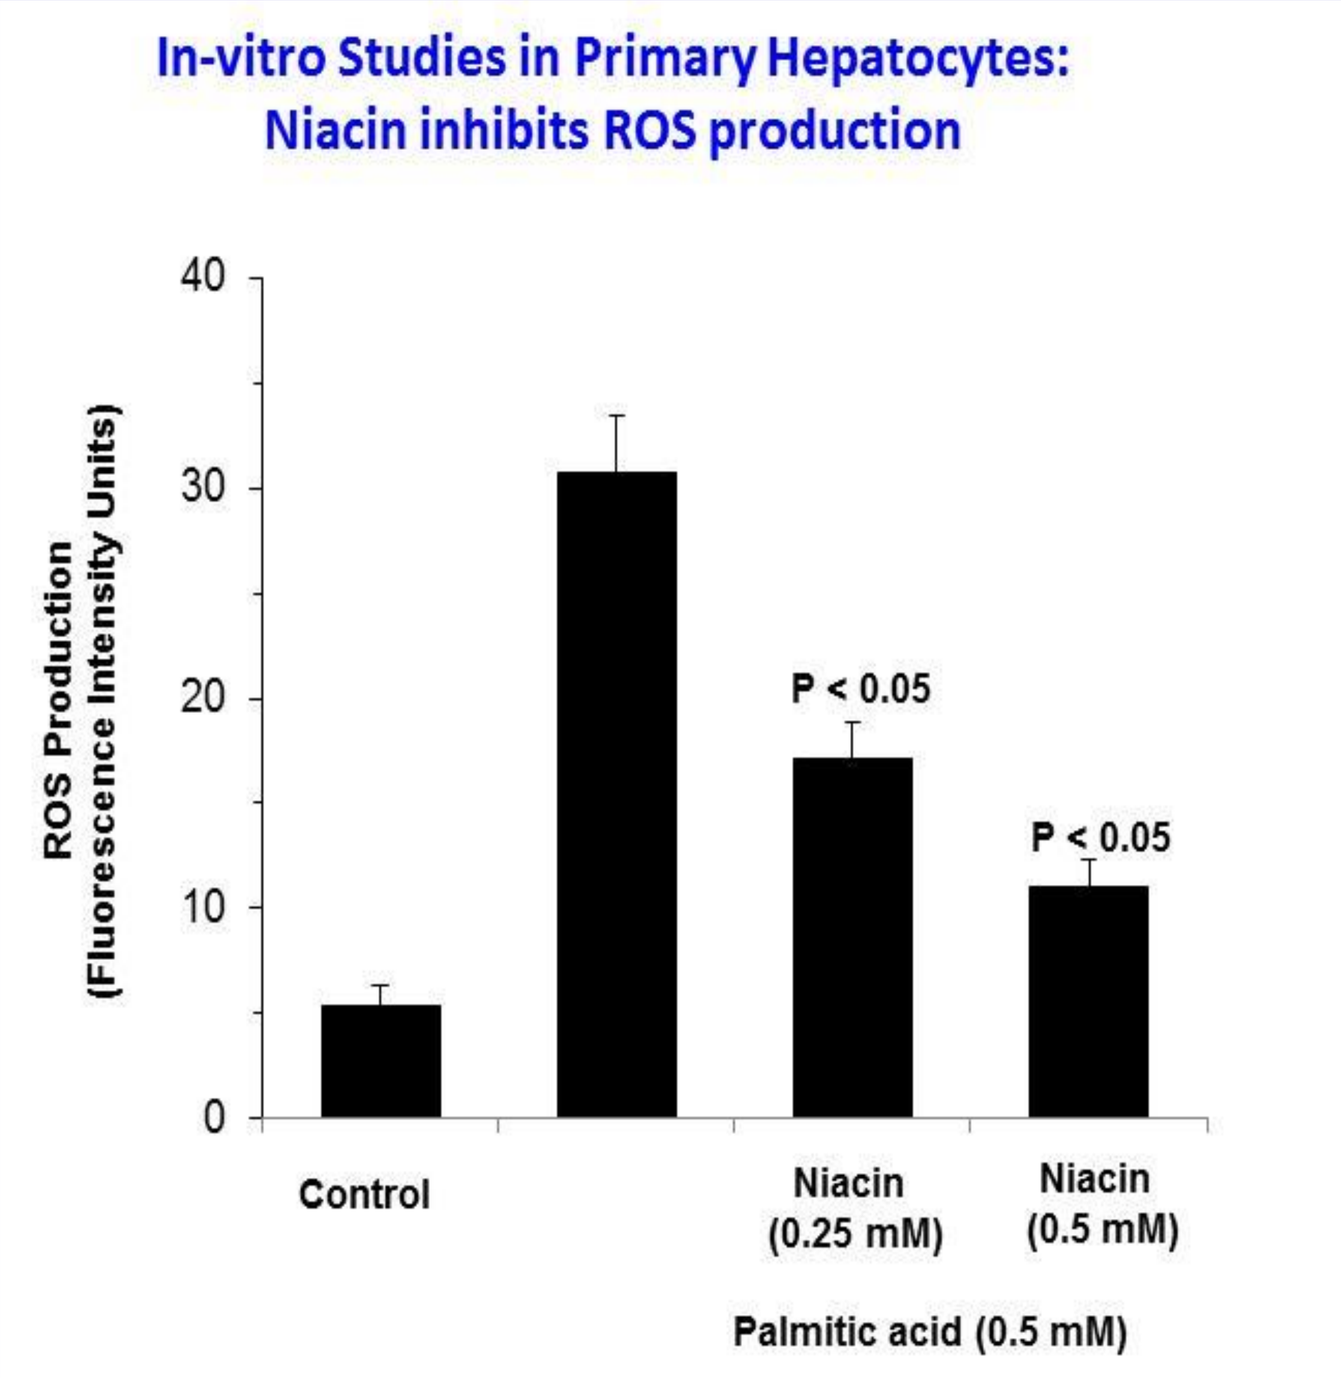
In-vitro Studies in Primary Hepatocytes: Niacin inhibits ROS production
ROS Production (Fluorescence Intensity Units)
P < 0.05
P < 0.05
Control
Niacin (0.25 mM)
Niacin (0.5 mM)
Palmitic acid (0.5 mM)

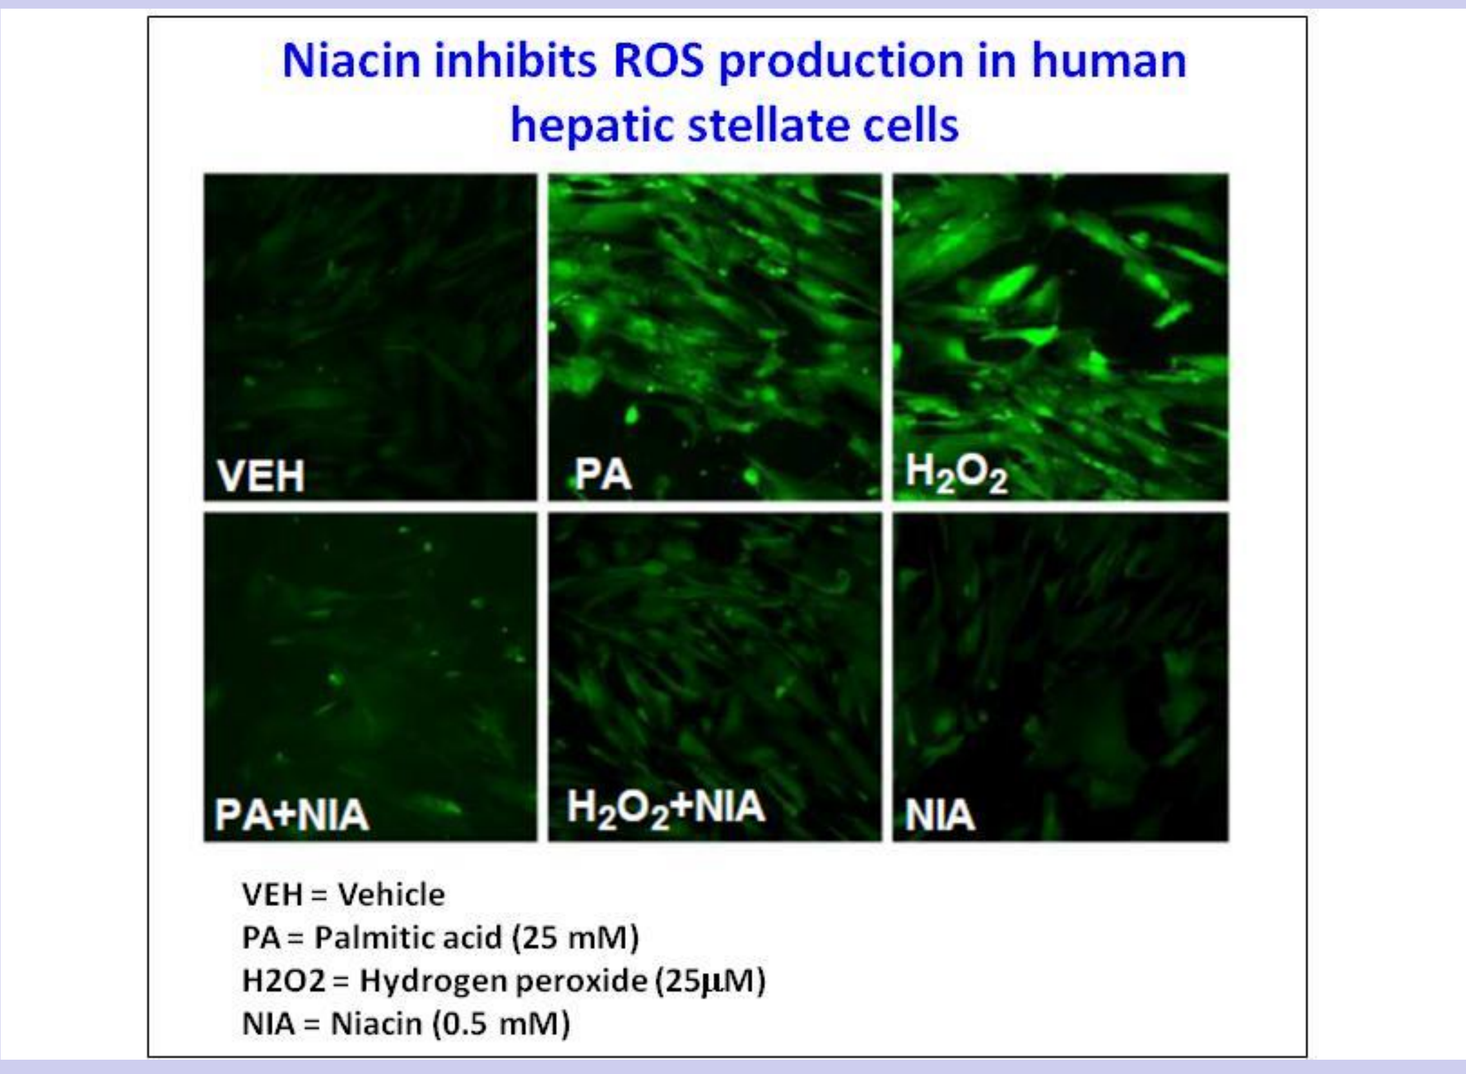
Niacin inhibits ROS production in human hepatic stellate cells
VEH
PA
H2O2
PA+NIA
H2O2+NIA
NIA
VEH = Vehicle
PA = Palmitic acid (25 mM)
H2O2 = Hydrogen peroxide (25µM)
NIA = Niacin (0.5 mM)

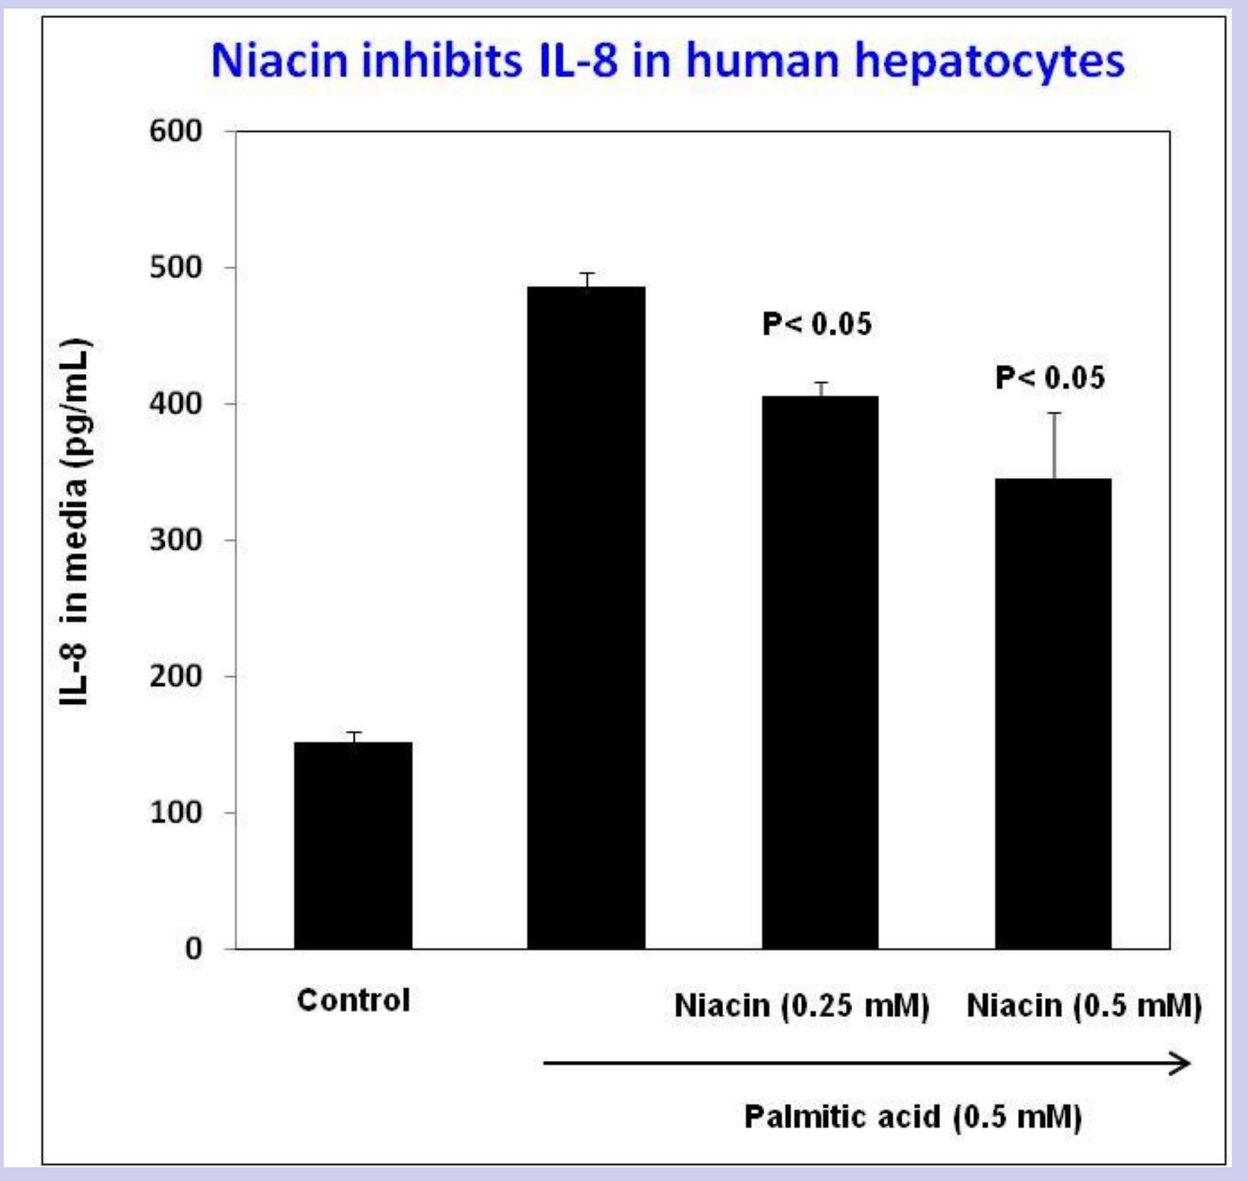
Niacin inhibits IL-8 in human hepatocytes
IL-8 in media (pg/mL)
P< 0.05
P< 0.05
Control
Niacin (0.25 mM)
Niacin (0.5 mM)
Palmitic acid (0.5 mM)

Ganji SH, et.al. Metabolism 64:980-990, 2015

Niacin Prevents and Regresses Fibrosis in Human Hepatic Stellate Cells from Normal Subjects and Patients with NASH

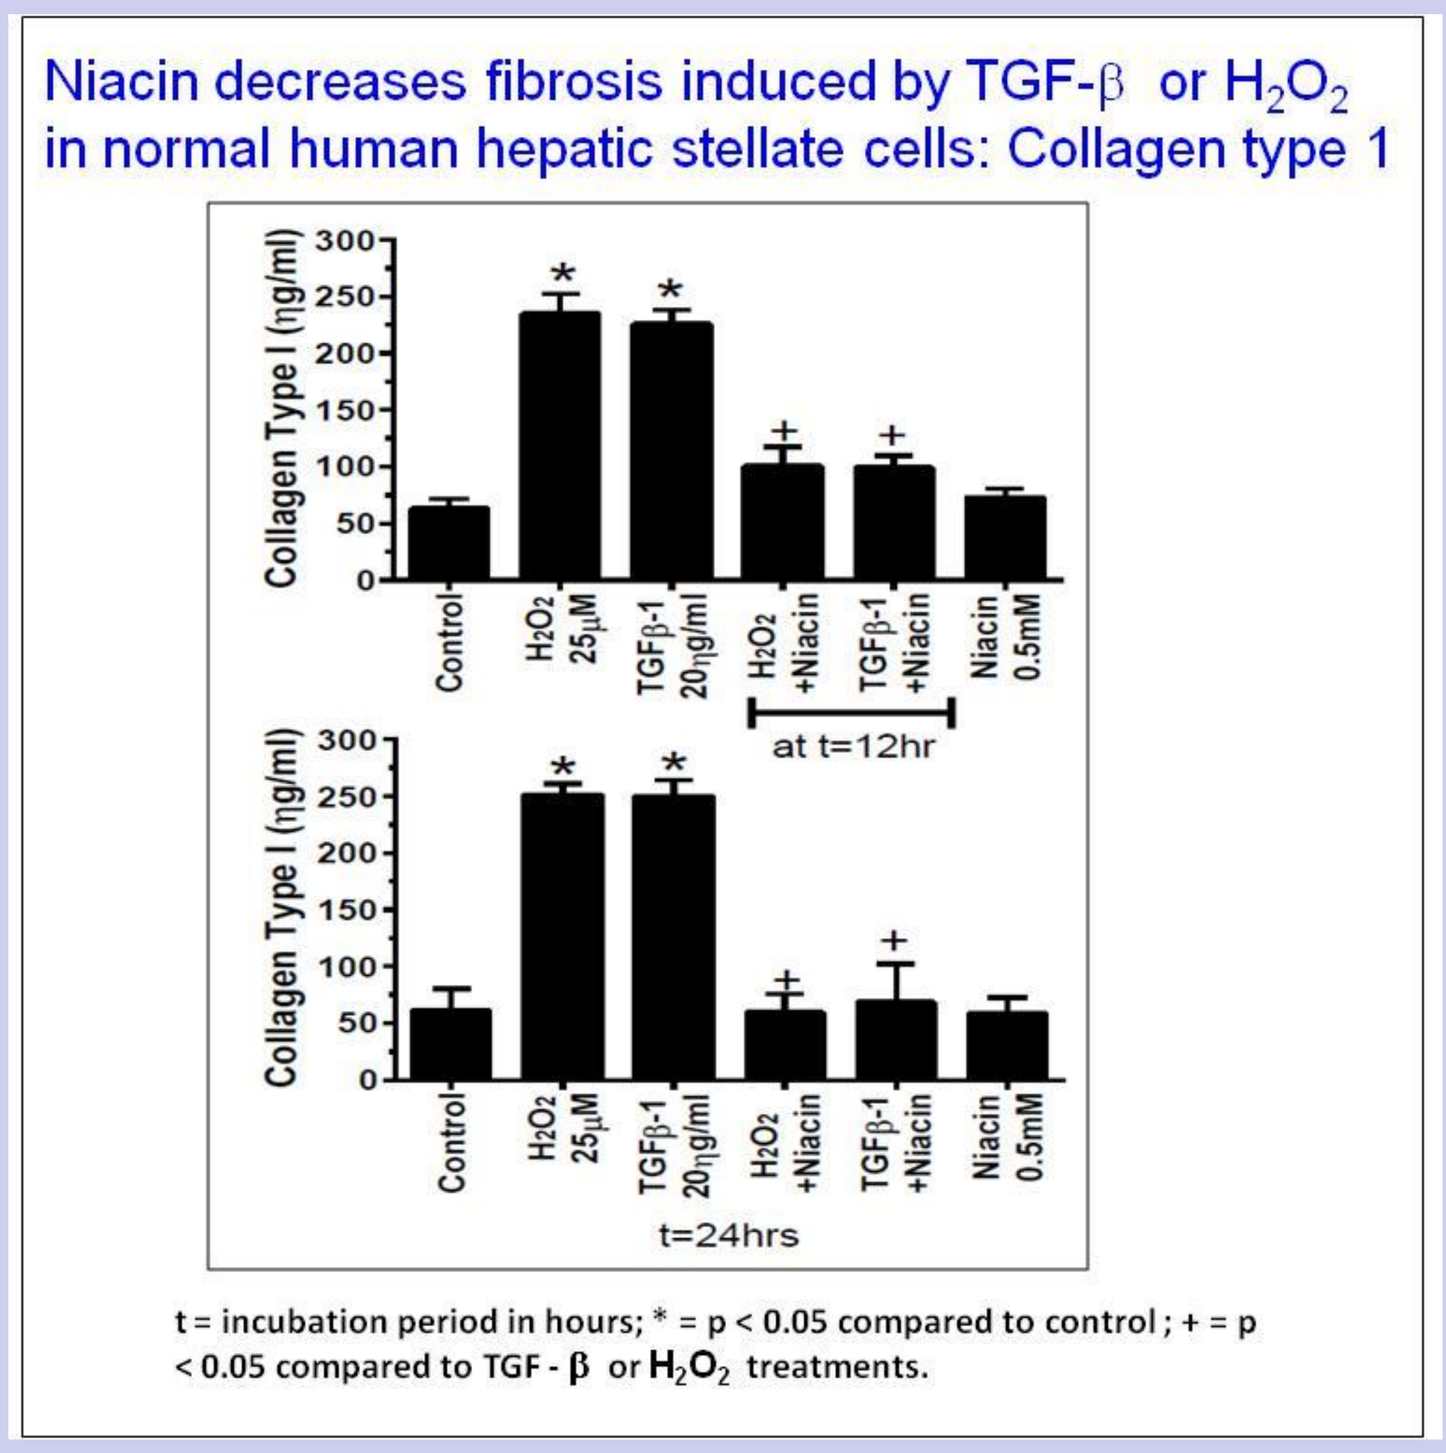
Niacin decreases fibrosis induced by TGF-β or H2O2 in normal human hepatic stellate cells: Collagen type 1
Collagen Type I (ng/ml)
Control
H2O2 25µM
TGFβ-1 20ηg/ml
H2O2 +Niacin
TGFβ-1 +Niacin
Niacin 0.5mM
at t=12hr
t=24hrs
t = incubation period in hours; * = p < 0.05 compared to control ; + = p < 0.05 compared to TGF - β or H2O2 treatments.

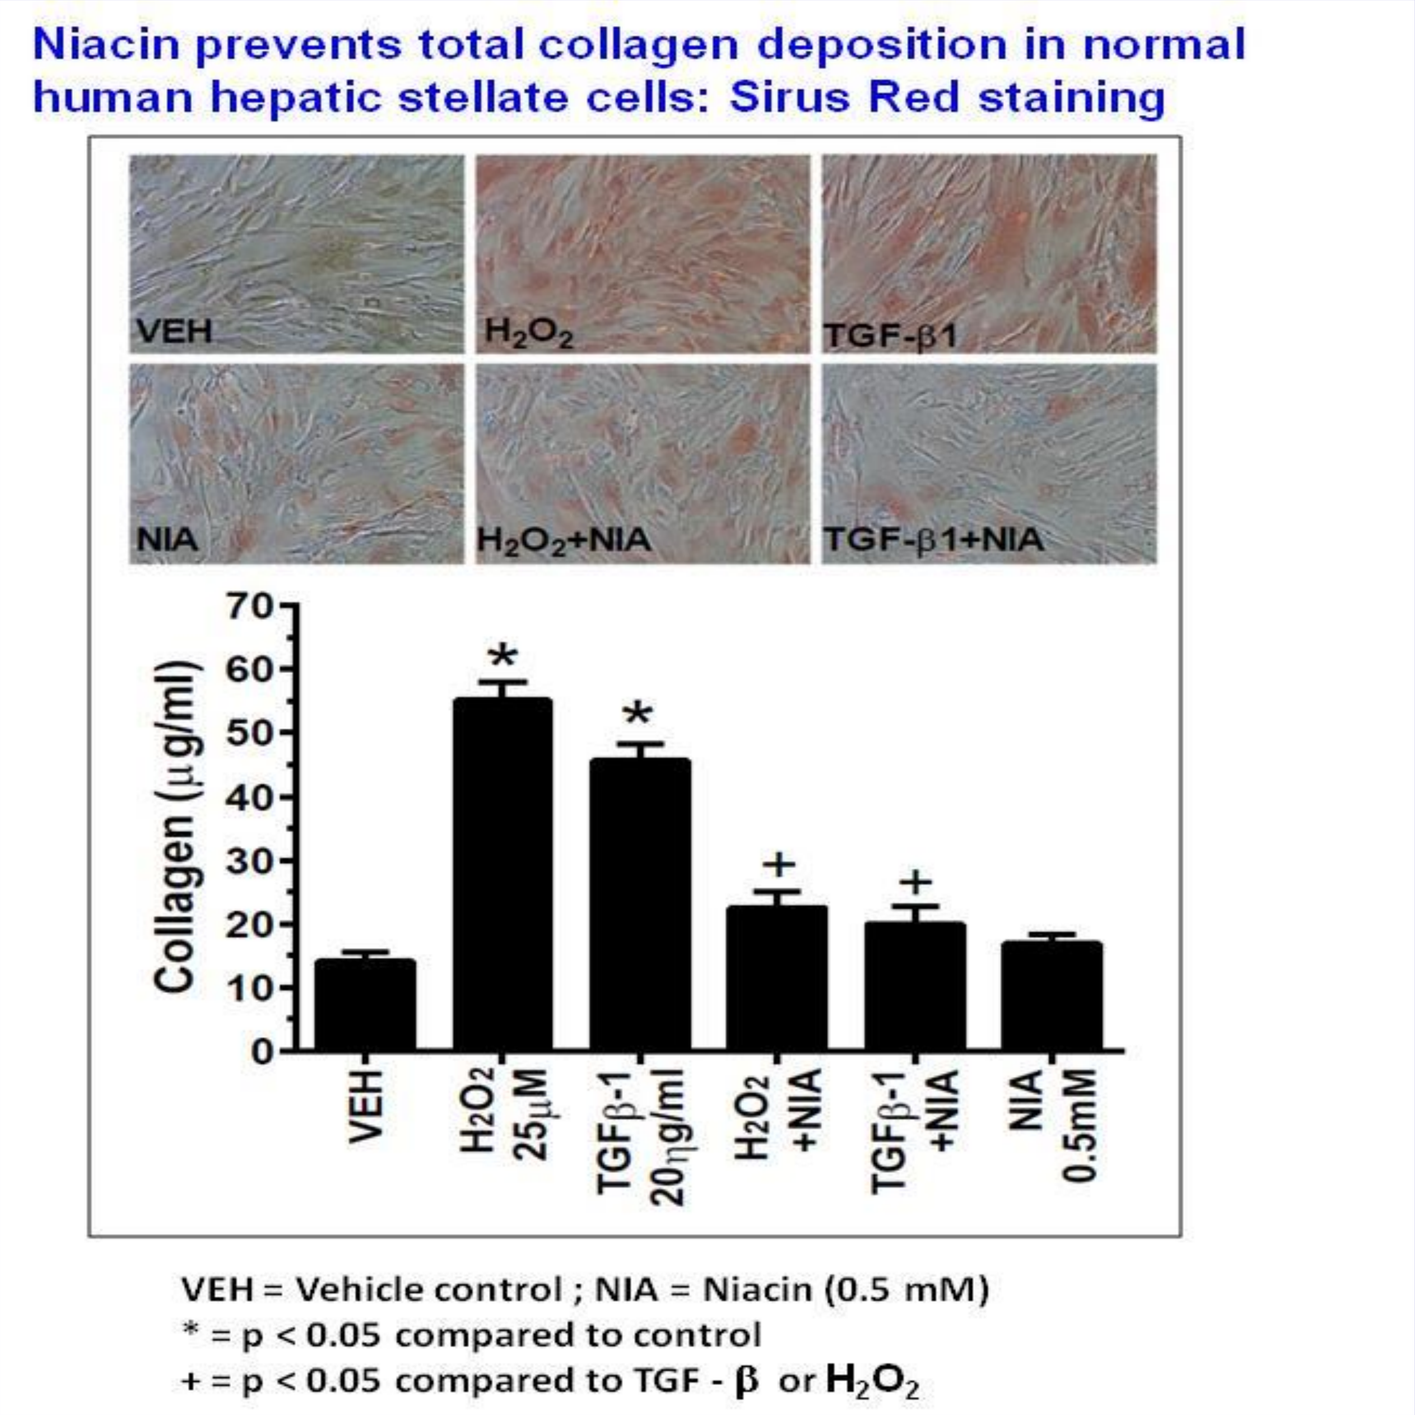
Niacin prevents total collagen deposition in normal human hepatic stellate cells: Sirus Red staining
VEH
H2O2
TGF-β1
NIA
H2O2+NIA
TGF-β1+NIA
Collagen (µg/ml)
VEH = Vehicle control ; NIA = Niacin (0.5 mM)
* = p < 0.05 compared to control
+ = p < 0.05 compared to TGF - β or H2O2

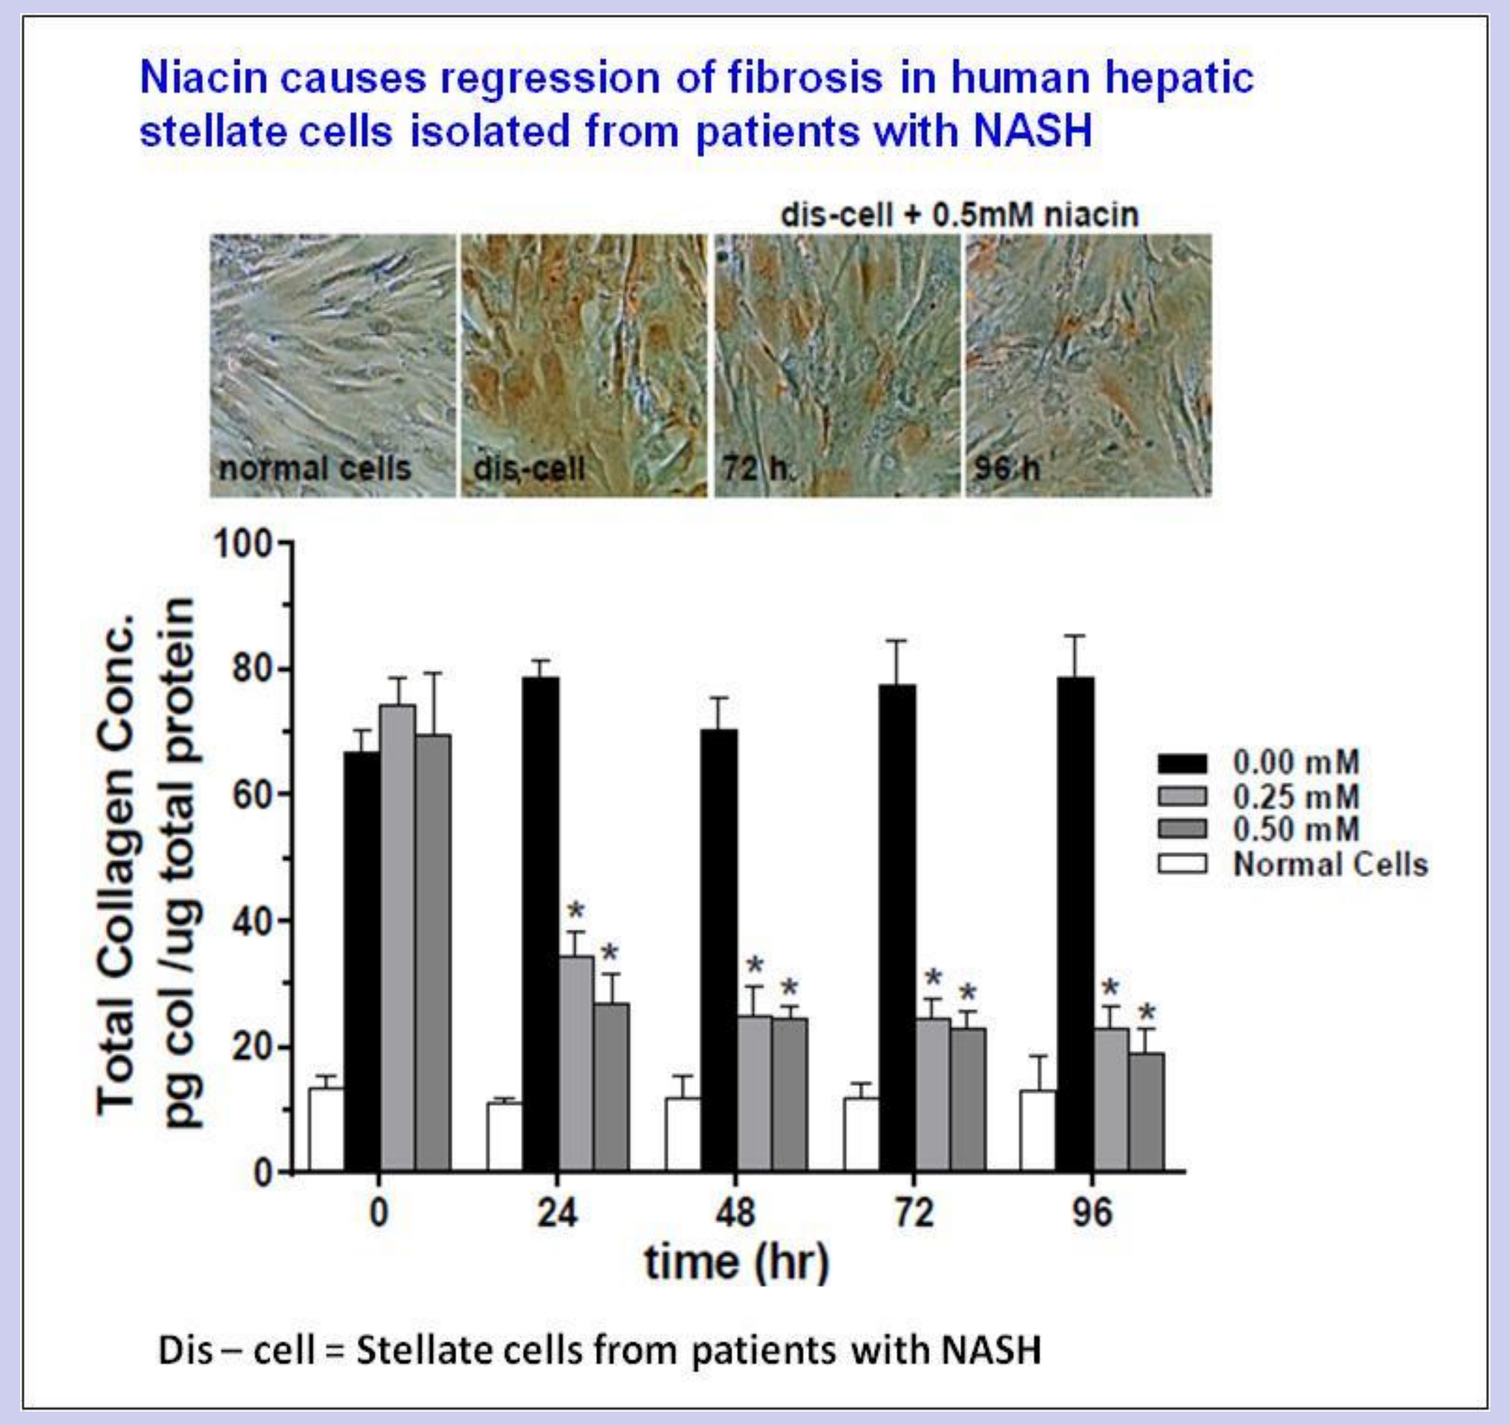
Niacin causes regression of fibrosis in human hepatic stellate cells isolated from patients with NASH
dis-cell + 0.5mM niacin
normal cells
dis-cell
72 h
96 h
Total Collagen Conc. pg col /ug total protein
time (hr)
0.00 mM
0.25 mM
0.50 mM
Normal Cells
Dis – cell = Stellate cells from patients with NASH

CONCLUSIONS

Because niacin acts on the 3 major stages of NASH-NAFLD, combination with a drug in development could result in a broader and more intense efficacy than either drug alone.

The novel repurposed patented use of Niacin for Fatty Liver Disease gives a tremendous time lead in drug development for NASH. The safety and efficacy for dyslipidemia (but not NASH) has been extensively documented for FDA approved generic, Extended Release Niacin which is available by prescription.

Niacin uniquely offers the potential of not only treating NASH but also concurrent dyslipidemia.

Niacin-based combination therapy research could produce one of the most effective *broad-spectrum* products for NASH.

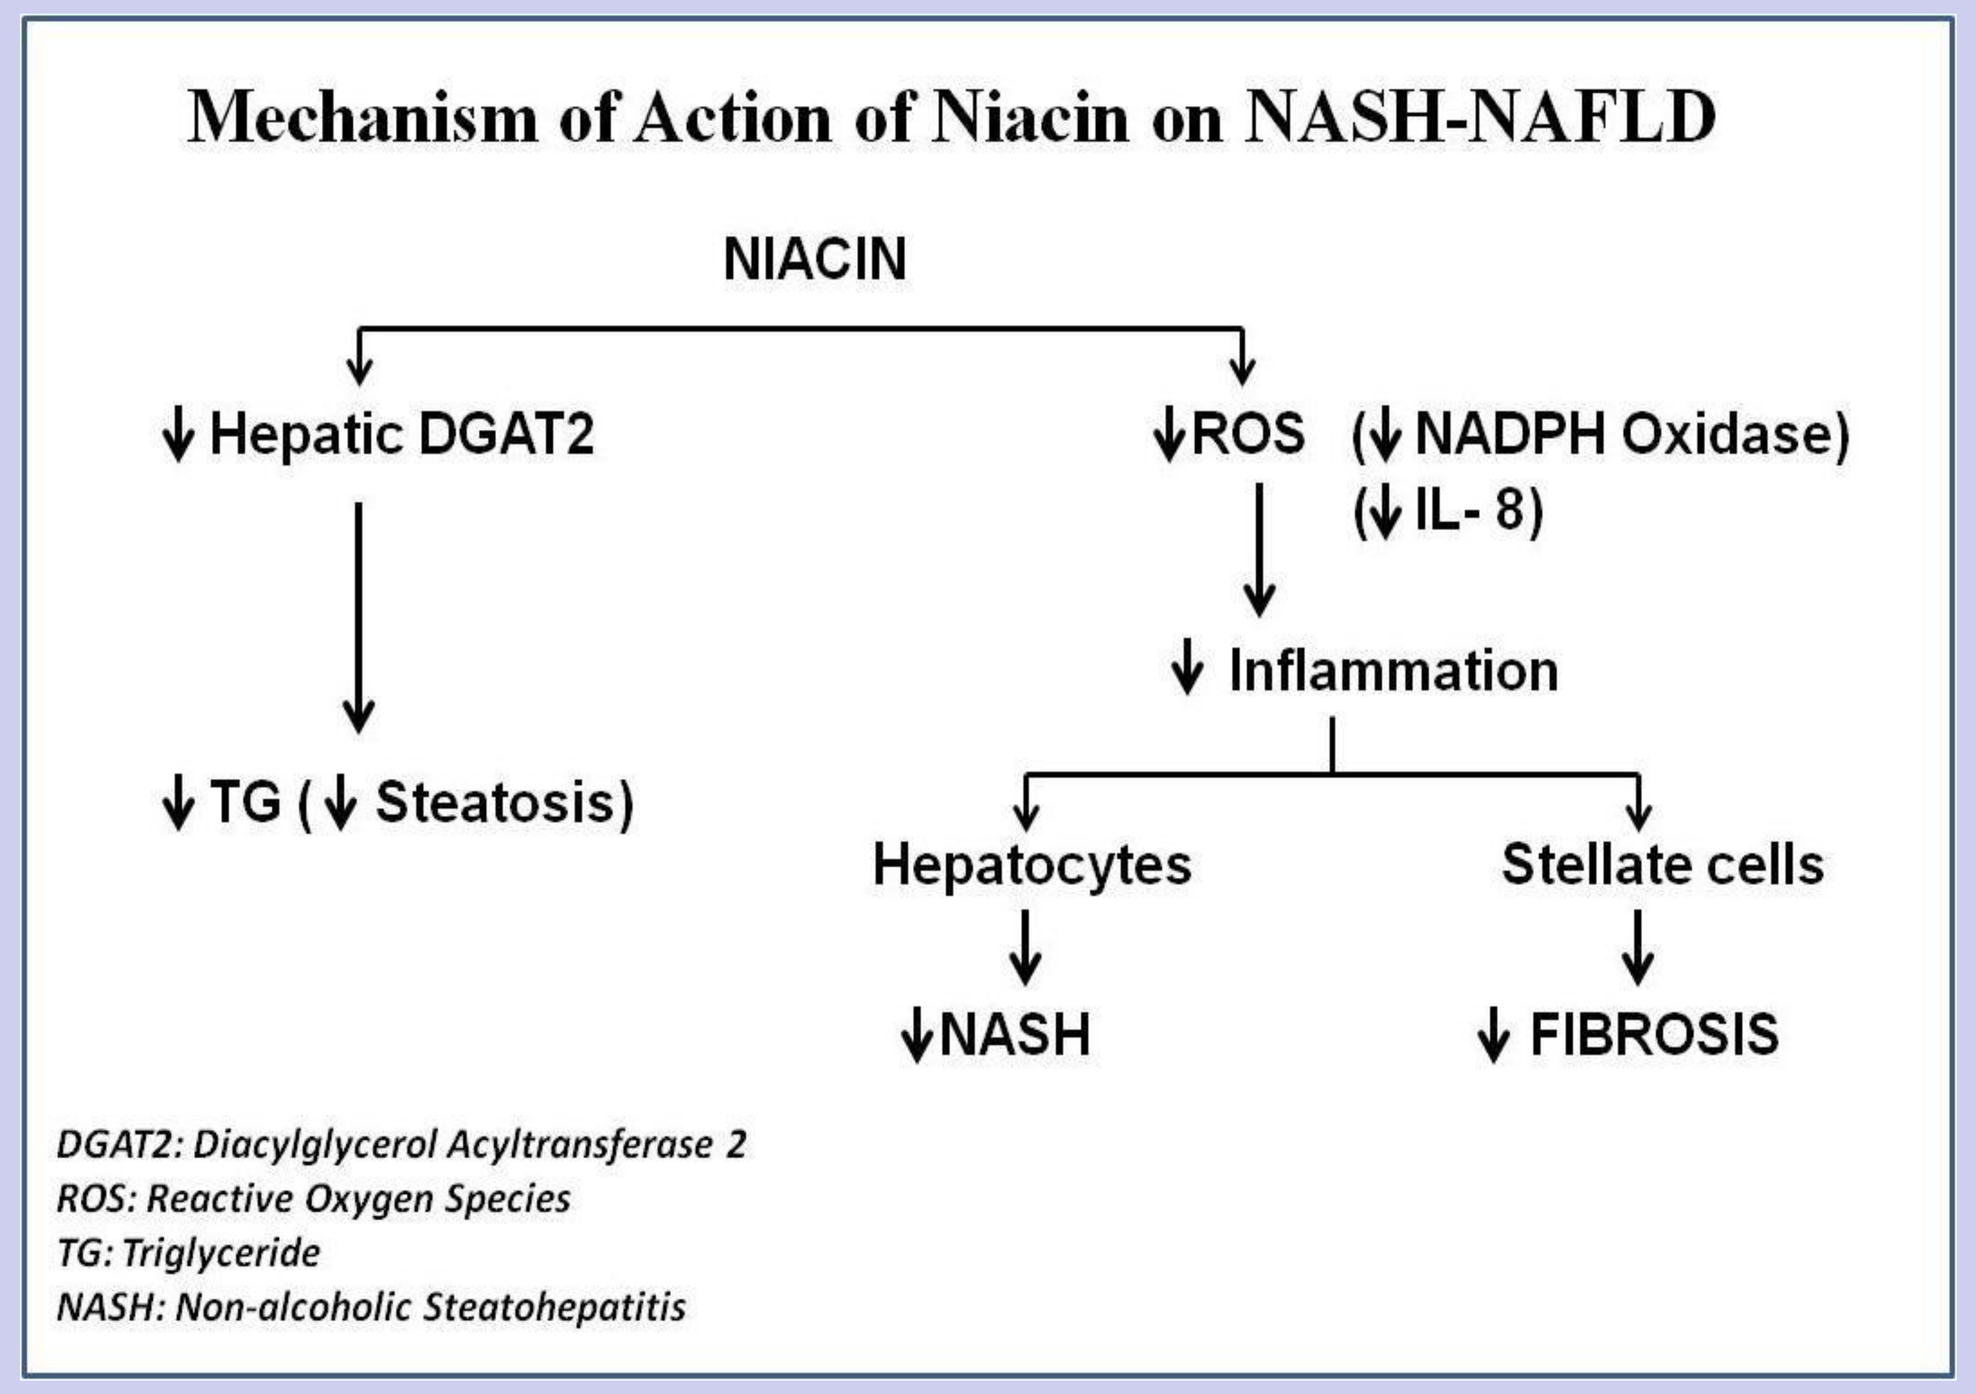
Mechanism of Action of Niacin on NASH-NAFLD
NIACIN
↓Hepatic DGAT2
↓ROS (↓NADPH Oxidase) (↓IL- 8)
↓ Inflammation
↓TG (↓ Steatosis)
Hepatocytes
Stellate cells
↓NASH
↓ FIBROSIS
DGAT2: Diacylglycerol Acyltransferase 2
ROS: Reactive Oxygen Species
TG: Triglyceride
NASH: Non-alcoholic Steatohepatitis

REFERENCES

1. Indication for use of niacin (nicotinic acid) for treatment and reversal of fatty liver disease. Patent no.: US 9, 072, 732 B2; July 7, 2015.
2. Angulo P. Nonalcoholic fatty liver disease. N. Eng. J. Med. 346:1221-1231, 2002.
3. Clark JM, Brancati FL, et al. Nonalcoholic fatty liver disease. Gastroenterol.122:1649-1657, 2002.
4. Oseini AM, Sanyal AJ. Therapies in non-alcoholic steatohepatitis (NASH). Liver Internat. 37:97-103, 2017.
5. Charlton MR, Burns JM, Pedersen RA, et. al. Frequency and outcomes of liver transplantation for nonalcoholic steatohepatitis in the United States. Gastroenterology 141:1249-1253, 2011.
6. Sanyal A, Poklepovic A, Moyneur E, et. al. Population-based risk factors and resource utilization for HCC: perspective. Curr. Med. Res. Opin. 26:2183-2179, 2010.